explicat® pharma GmbH
Development and Consulting

FRIEDRICH-ALEXANDER
UNIVERSITÄT
ERLANGEN-NÜRNBERG
FACULTY OF SCIENCES

A CASE STUDY TO OPTIMIZE FREEZE DRYING CONDITIONS OF CYCLODEXTRIN-BASED FORMULATIONS

Martina Schuller [1,2], Andrea Weiland [2], Henning Gieseler [1]
[1] Friedrich-Alexander-University (FAU) Erlagen-Nürnberg, Department of Pharmaceutics, Cauerstrasse 4, 91058 Erlangen
[2] Explicat Pharma GmbH, Georg-Knorr-Straße 4, 85662 Hohenbrunn, Germany
Corresponding author: Martina Schuller, m.schuller@explicat.com , phone + 49 8102 72785 26

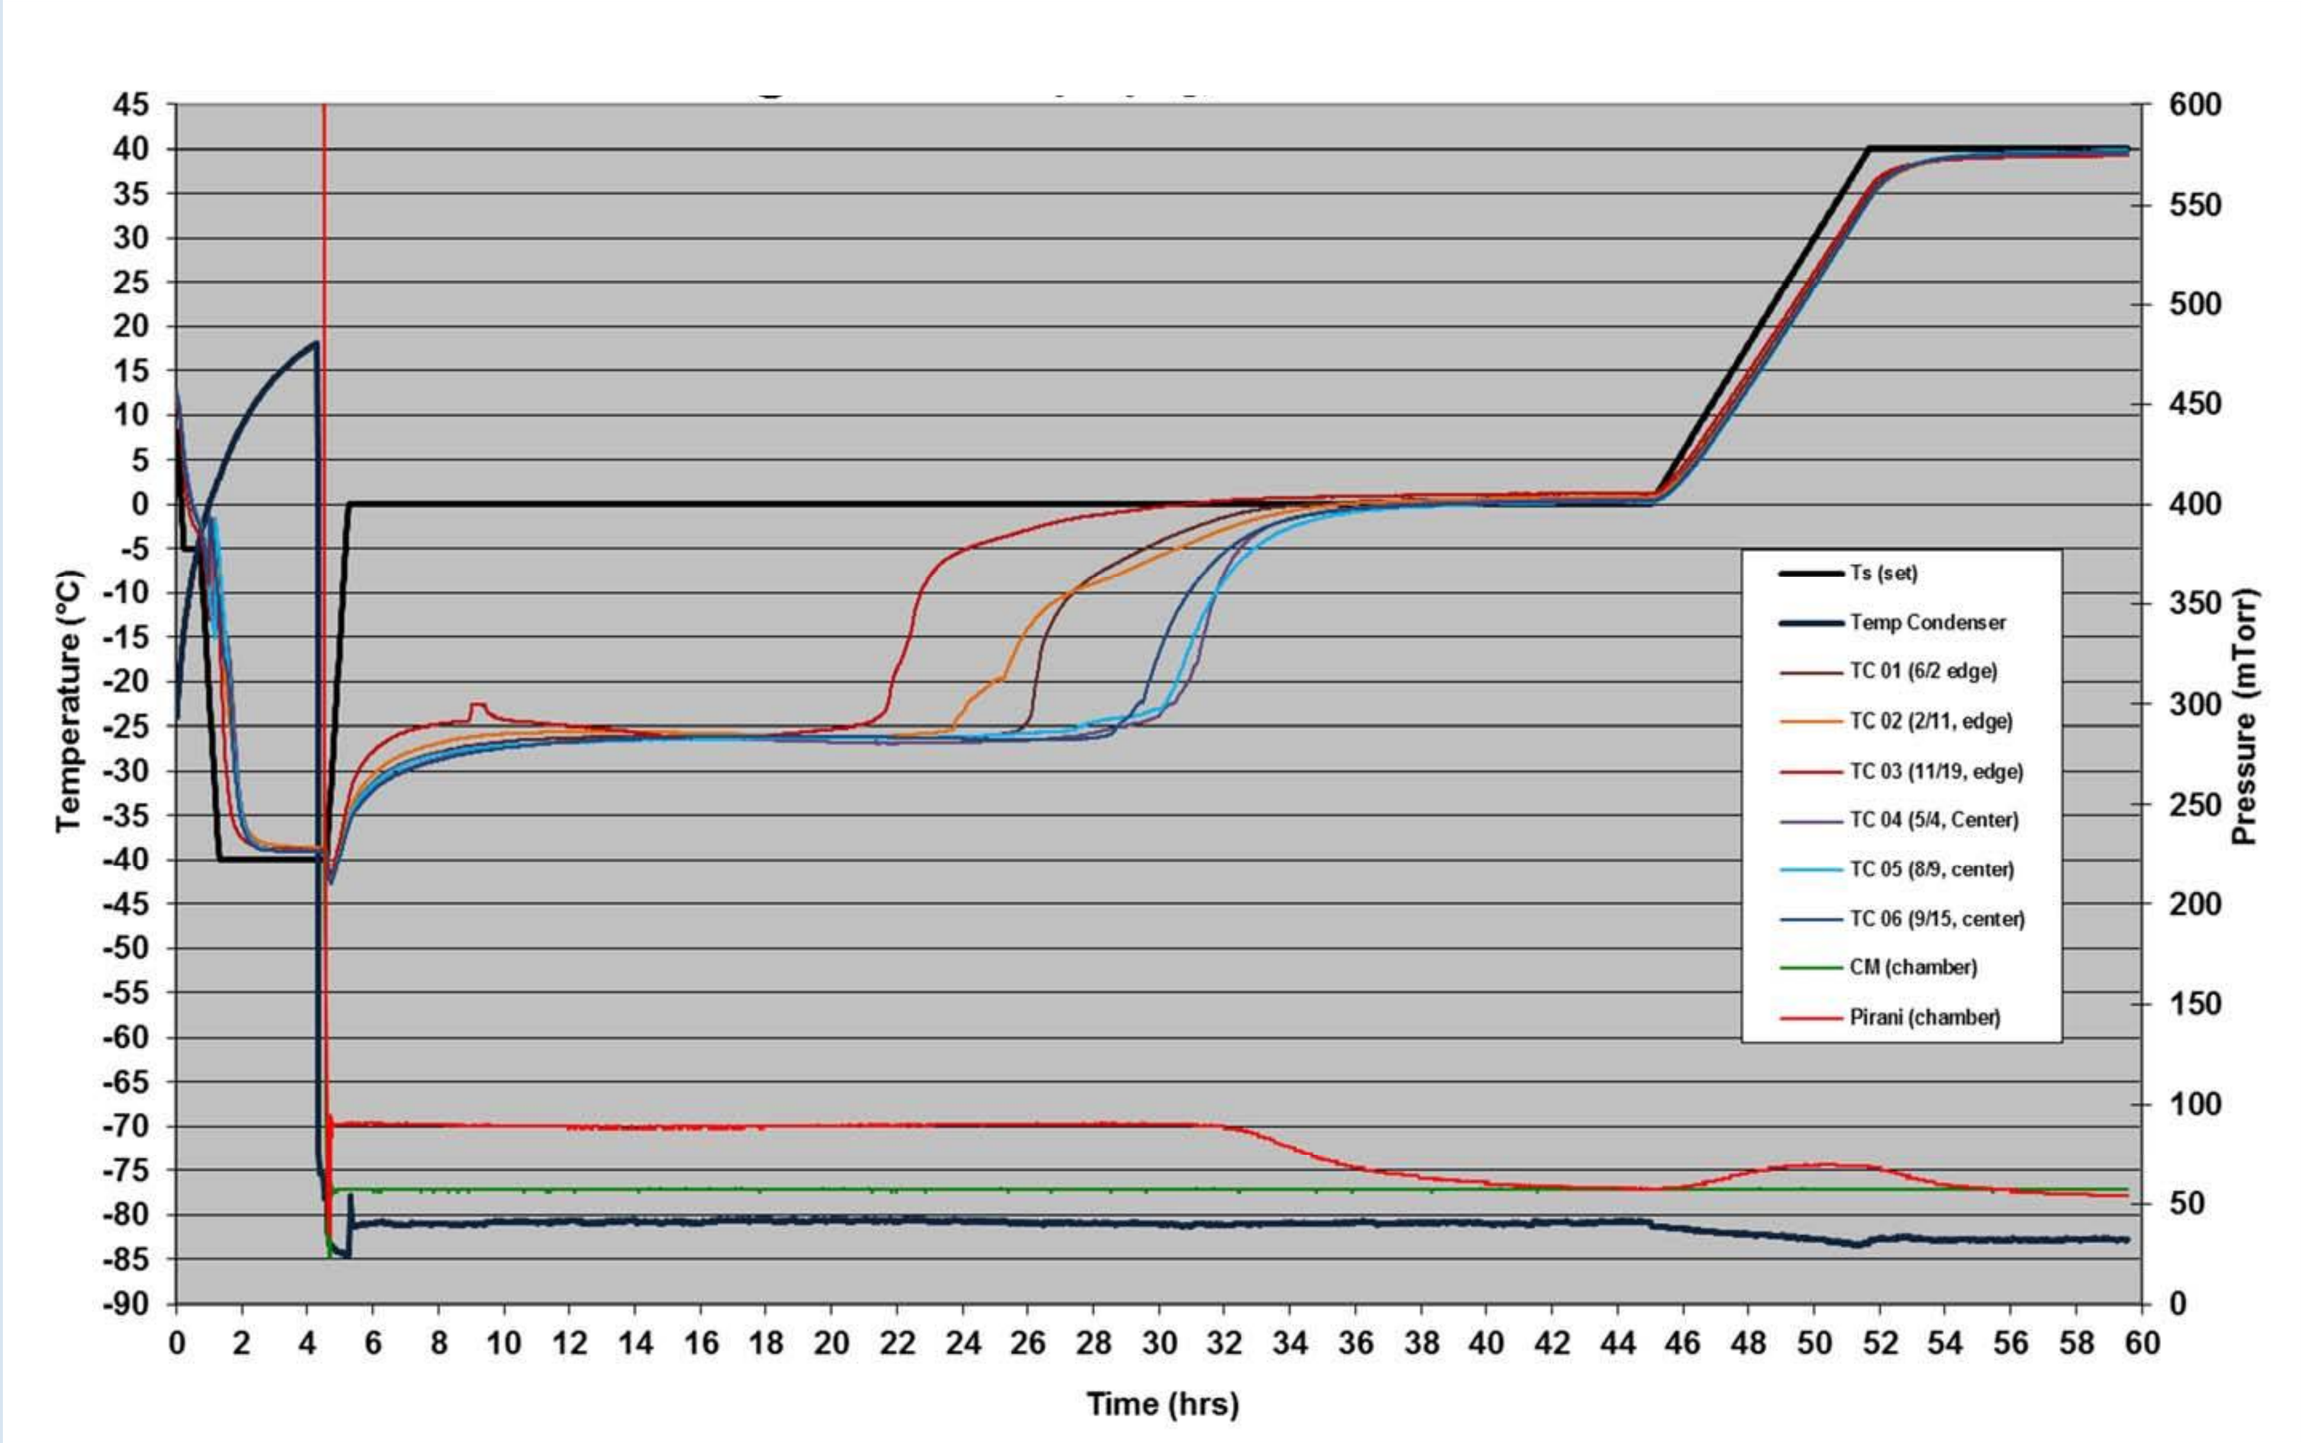
Temperature (°C)
Pressure (mTorr)
Time (hrs)
Ts (set)
Temp Condenser
TC 01 (6/2 edge)
TC 02 (2/11, edge)
TC 03 (11/19, edge)
TC 04 (5/4, Center)
TC 05 (8/9, center)
TC 06 (9/15, center)
CM (chamber)
Pirani (chamber)
Figure 1: Placebo run without annealing step – primary drying at Ts = 0°C, secondary drying at Ts = 40°C for 8 h, partial load

INTRODUCTION

SBECD (Sulfobutylether - β - Cyclodextrin) is a new and promising excipient for freeze dried formulations. The present study illustrates an approach to optimize freeze drying conditions for formulations containing SBECD.

First, a placebo was used to evaluate a general freeze drying recipe. Further, the information gained was compared with drug product runs.

MATERIALS AND METHODS

Formulation composition: SBECD, WFI, NaOH for pH adjustment, proprietary small molecule API

Freeze dryer: 0.5 m^2 pilot scale freeze dryer (LyoStar, SP Scientific, Gardiner, NY, USA)

Product presentation: 10R vials filled with 5 mL; igloo stopper, bromobutyl, siliconized

Instrumentation: 36 gauge thermocouples (TC); comparative pressure measurement

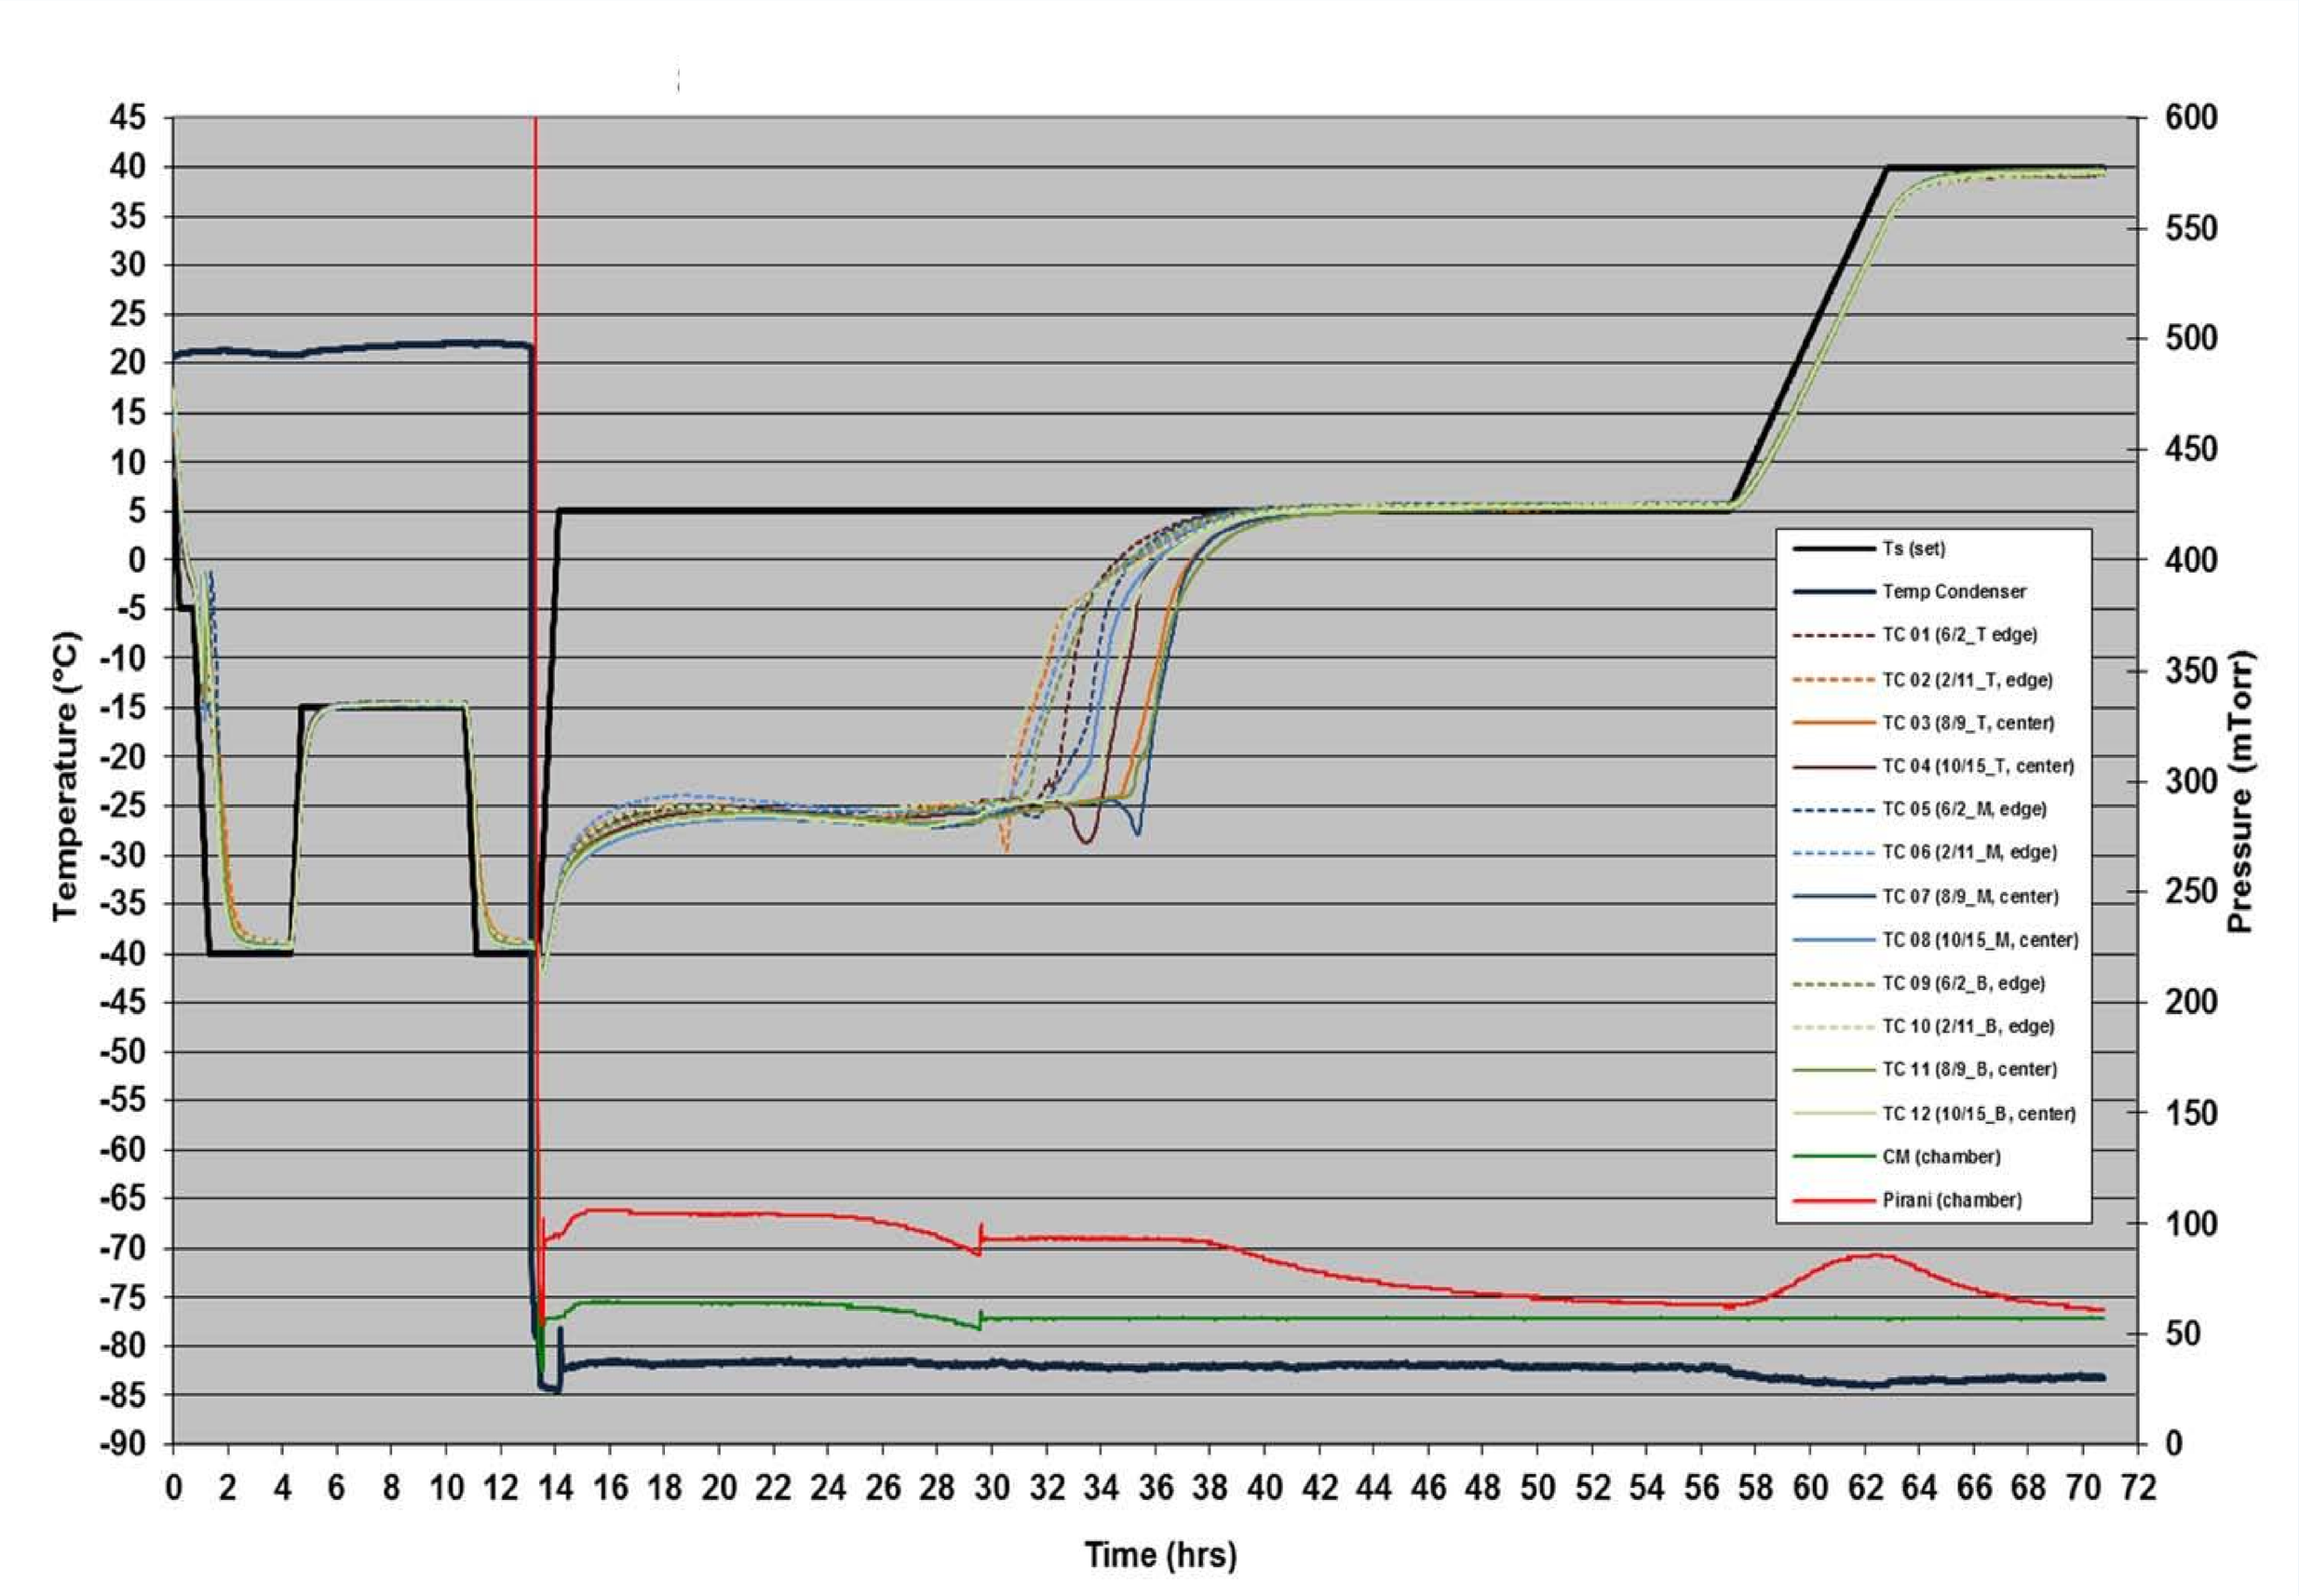
Temperature (°C)
Pressure (mTorr)
Time (hrs)
Ts (set)
Temp Condenser
TC 01 (6/2_T edge)
TC 02 (2/11_T, edge)
TC 03 (8/9_T, center)
TC 04 (10/15_T, center)
TC 05 (6/2_M, edge)
TC 06 (2/11_M, edge)
TC 07 (8/9_M, center)
TC 08 (10/15_M, center)
TC 09 (6/2_B, edge)
TC 10 (2/11_B, edge)
TC 11 (8/9_B, center)
TC 12 (10/15_B, center)
CM (chamber)
Pirani (chamber)
Figure 2: Drug product run with annealing step – primary drying at Ts = 5°C, secondary drying at Ts = 40°C for 8 h, full load

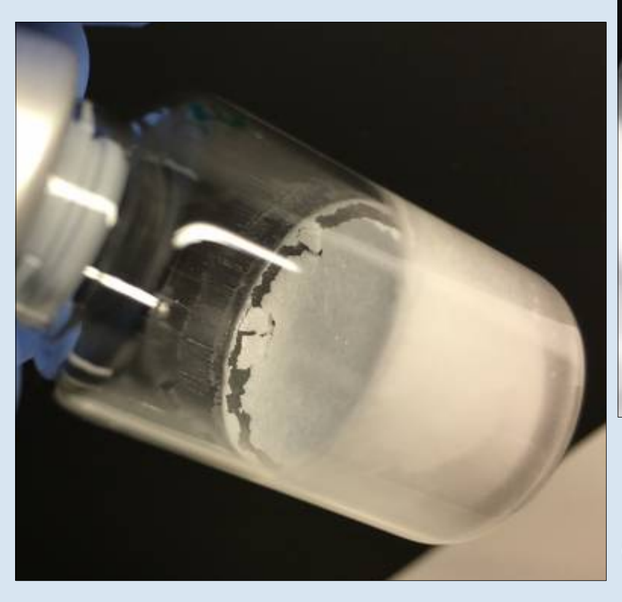
Figure 3: Shrinkage

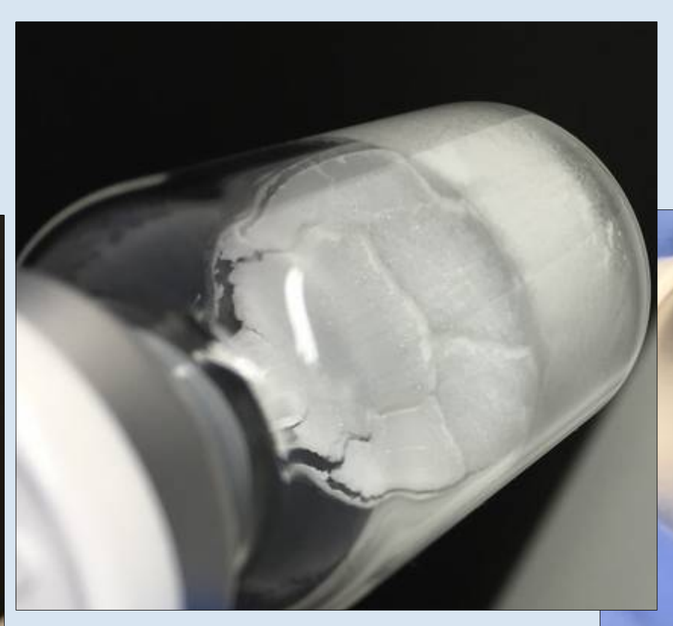
Figure 4: Cracking

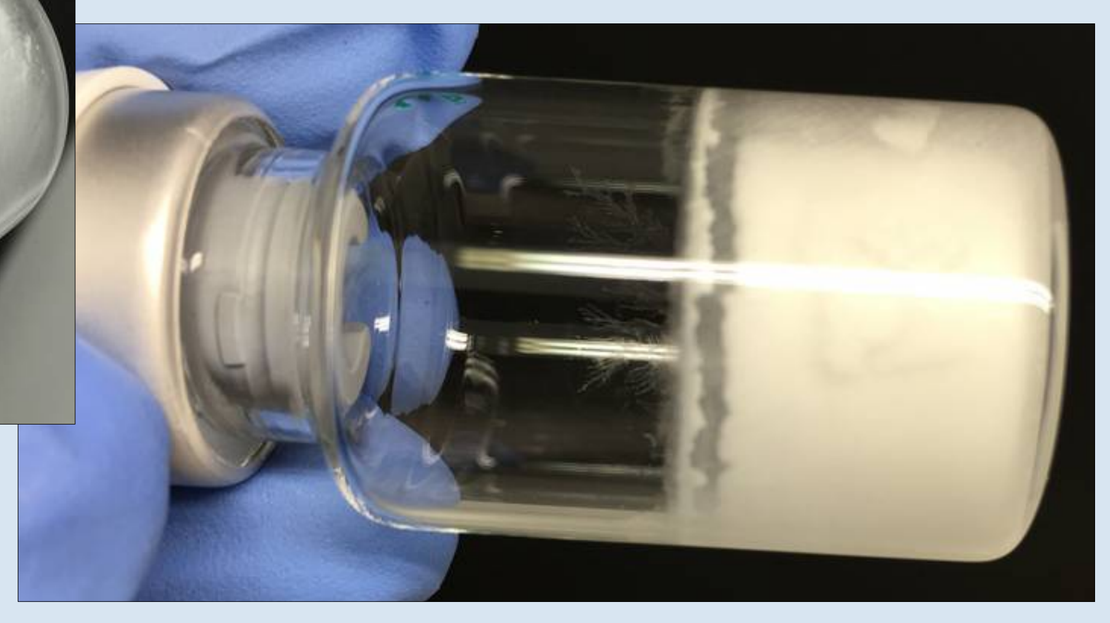
Figure 5: Fogging

RESULTS

Placebo runs led only to cosmetic and thus acceptable defects: crack formation, shrinkage and fogging. Through introduction of an annealing step the tendency of cracking could be minimized.

Product temperature profiles suggested that SBECD as excipient would allow more rigorous process conditions in freeze drying. Therefore, shelf temperature was raised and primary drying could be reduced by approximately 35 %.

Introduction of a proprietary small molecule API led to similar product and process performance attributes than the corresponding placebo. The molar ratio of API to SBECD of 1:9 contributed positively to this result.

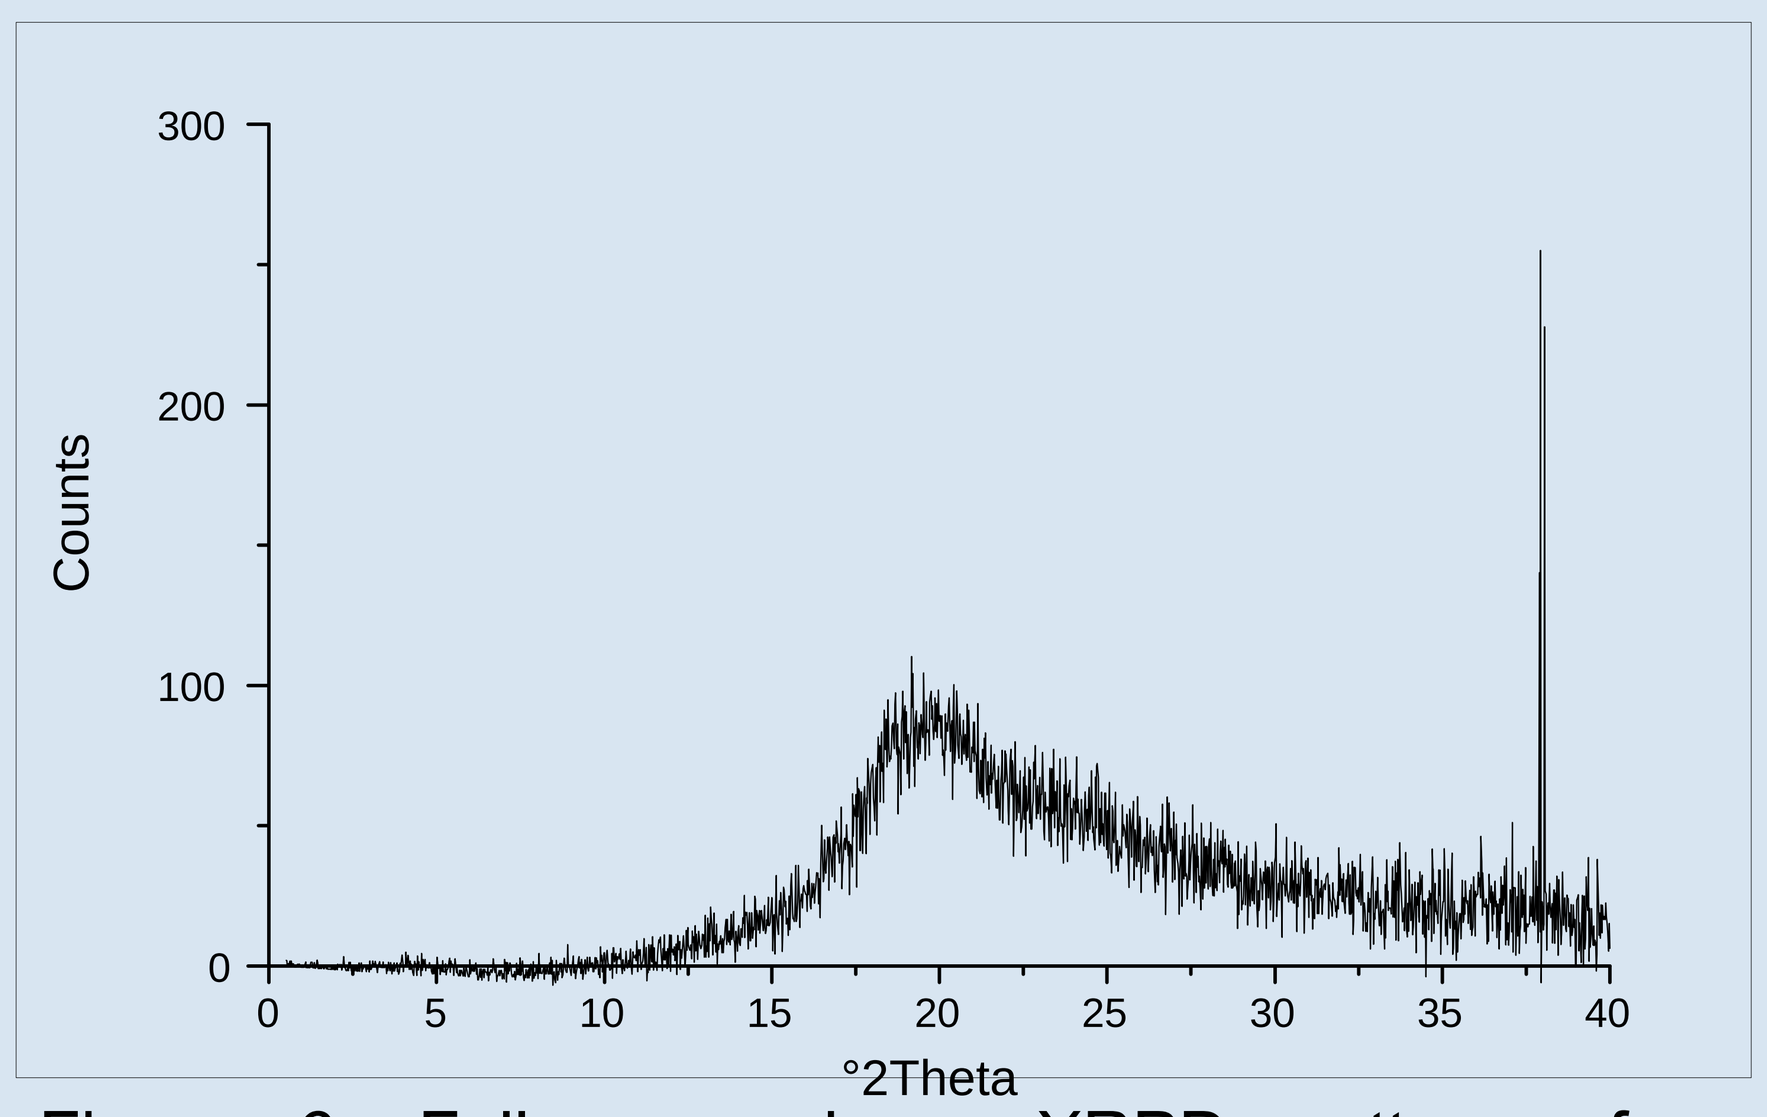
Counts
°2Theta
Figure 6: Fully amorphous XRPD pattern of a lyophilizate containing SBECD and API

CONCLUSION

SBECD is an excipient which provides the advantage of using aggressive cycle conditions during freeze drying. Comparably aggressive conditions can only be found for mannitol-based formulation systems.

Table 1:
Mean water content in the lyophilizate after placebo runs with varying parameters (evaluated by Karl-Fischer titration - Ph. Eur. 2.5.12)

Ts = 0°C, no annealing, 1 shelf, 60 h of total process time	1.61 %
Ts = 0°C, annealing for 6 h at -15°C, 1 shelf, 75 h of total process time	1.57 %
Ts = 10°C, annealing for 6 h at -15°C, 1 shelf, 56 h of total process time	1.41 %

REFERENCES
(1) Searles et al.: "Annealing to Optimize the Primary Drying Rate, Reduce Freezing-Induced Drying Rate Heterogeneity, and Determine Tg' in Pharmaceutical Lyophilization", in: J. Pharm. Sci., 2001 Jul; 90(7): 872-887.
(2) Geidobler, Raimund Michael: Cyclodextrins as Excipients in Drying of Proteins and Controlled Ice Nucleation in Freeze-drying, Diss. LMU München 2014, p. 61.
(3) Rambhatla et al.: "Cake Shrinkage During Freeze Drying: A Combined Experimental and Theoretical Study", in: Pharm. Dev. Technol., 2005, p. 33 - 40.
(4) Patel et al.: "Lyophilized Drug Product Cake Appearance: What is Acceptable?", in. J.Pharm.Sci., 2017 Jul; 106(7): 1706-1721.
(5) Patel et al.: "Determination of End Point of Primary Drying in Freeze-Drying Process Control", in: AAPS PharmSciTech, 2010 Mar; 11(1): 73-84.